Poster P5-19-12

BRAWO — BReast Cancer with Afinitor ER+ WOmen

Subgroup Analysis of Efficacy in Routine Treatment—Results of the 2nd Interim Analysis of BRAWO, the Non-Interventional Trial "Breast Cancer Treatment With Everolimus and Exemestane for HR$^+$ Women"

Christian Jackisch,[1] Eva-Maria Grischke,[2] Andreas Schneeweiss,[3] Thomas Decker,[4] Christoph Uleer,[5] Frank Förster,[6] Oliver Tomé,[7] Pauline Wimberger,[8] Christian Kurbacher,[9] Bettina Mueller,[10] Nadia Harbeck,[11] Christoph Mundhenke,[12] Sherko Kümmel,[13] Mathias Muth,[14] Julia Kreuzeder,[14] Wilhelm Bloch,[15] Hans Tesch,[16] Diana Lueftner,[17] Florian Schütz,[3] Peter Fasching[18]

[1]Breast Center at the Hospital Offenbach, Offenbach, Germany; [2]Department of Obstetrics and Gynecology, University Tuebingen, Tuebingen, Germany; [3]Department of Obstetrics and Gynecology, University Heidelberg, Heidelberg, Germany; [4]Medical Center Ravensburg, Ravensburg, Germany; [5]Medical Center Hildesheim, Hildesheim, Germany; [6]Poliklinik GmbH Chemnitz, Chemnitz, Germany; [7]Women's Hospital and School for Midwives, St. Vincentius Clinic, Karlsruhe, Germany; [8]Department of Obstetrics and Gynecology, Technical University, Dresden, Dresden, Germany; [9]Gynecology and Obstetrics, Medical Center Bonn-Friedensplatz, Bonn, Germany; [10]Obstetrics & Gynecology, The GRN Health Centers, Rhein-Neckar, Weinheim, Germany; [11]Department of Obstetrics and Gynecology, Hospitals Grosshadern and Maistrasse-Innenstadt, University of Munich, Munich, Germany; [12]Department of Obstetrics and Gynecology, University Hospital of Schleswig-Holstein, University of Kiel, Kiel, Germany; [13]Interdisciplinary Breast Center, The Kliniken Essen-Mitte, University of Duisburg, Essen, Germany; [14]Novartis Pharma GmbH, Nuremberg, Germany; [15]Department of Sport Medicine, University of Cologne, Cologne, Germany; [16]Oncology Bethanien, Frankfurt/Main, Germany; [17]Department of Hematology, Oncology, and Tumor Immunology, Charité University Medicine, Campus Benjamin Franklin, Berlin, Germany; [18]Department of Obstetrics and Gynecology, University of Erlangen, Erlangen, Germany

Introduction

- In the pivotal phase 3 BOLERO-2 trial, the combination of everolimus (EVE) with exemestane (EXE) more than doubled median progression-free survival (PFS) versus placebo (PBO)+EXE in postmenopausal women with hormone-receptor-positive (HR$^+$) advanced breast cancer progressing after a nonsteroidal aromatase inhibitor[1]
 - 7.8 months with EVE+EXE versus 3.2 months with PBO+EXE (hazard ratio [HR]= 0.45; $P < .0001$) by local radiologic assessment
 - Results were confirmed by central radiologic assessment (11.0 months vs 4.1 months, respectively; HR = 0.38; $P < .0001$)
- BRAWO is a large, German, non-interventional study (NIS) with a planned enrollment of 3,000 patients with HR$^+$ advanced breast cancer receiving EVE+EXE
- Main objectives of the BRAWO study are to extend knowledge in the following areas
 - Efficacy, and the impact of physical activity on efficacy and quality of life, in routine clinical care
 - Prophylaxis and management of stomatitis
 - Sequence of therapy and drug utilization when EVE+EXE is used in routine clinical care
- In this second, preplanned, interim analysis, we present efficacy data for EVE+EXE in real-world conditions

Methods

- BRAWO is a large, German, NIS with data collected at ~400 sites (**Figure 1**)
- All patients with HR$^+$ advanced breast cancer receiving EVE+EXE treated in accordance with EVE labeling text are eligible for inclusion in BRAWO (planned N = 3,000)
 - The BRAWO trial population will be broader than the BOLERO-2 study population (eg, previous EXE therapy and more than 1 previous chemotherapy in the palliative setting are allowed)

Figure 1. BRAWO study design and objectives.

- Postmenopausal women
- HR$^+$, HER2$^-$ advanced breast cancer
- No symptomatic visceral metastasis
- Disease refractory to nonsteroidal aromatase inhibitors
- Treatment with EVE+EXE according to the clinical routine and labeling text from the EVE SmPC

Approximately 3,000 patients at 400 sites

- Non-interventional study
- Patients observed for the duration of treatment with EVE+EXE
- Observation intervals after initiating EVE+EXE:
 - 2 weeks
 - 1 month
 - 3 months
 - Every 3 months thereafter
- Observation ends no later than 1 year after last patient enrolled

Start of enrollment: OCT 2012; End of enrollment: DEC 2015; End of documentation: DEC 2016

Primary Objective:
- Efficacy (PFS) and impact of physical activity on efficacy (PFS)

Secondary Objectives:
- Quality of life and impact of physical activity on quality of life
- Overall response rate (ORR; complete response [CR] plus partial response [PR]) as assessed by investigator evaluation
- Stomatitis management—prophylactic measures and treatment of stomatitis in clinical routine
- Sequence of therapies and utilization of EVE in clinical routine

Abbreviations: EVE, everolimus; EXE, exemestane; HER2$^-$, human epidermal growth factor receptor-2–negative; HR$^+$, hormone receptor-positive; PFS, progression-free survival; SmPC, summary of product characteristics.
Study number: CRAD001JDE53

- The planned individual observation period per patient corresponds with the duration of EVE+EXE treatment, but ends no later than at the formal study end (1 year after the last patient enters the trial)
- The observation intervals are in line with clinical practice (ie, after approximately 2 weeks, 1 and 3 months, and thereafter at 3-month intervals after the start of treatment with EVE+EXE)
- Quality of life and physical activity are assessed by patient questionnaires:
 - European Organisation for Research and Treatment of Cancer Quality-of-Life Questionnaire-Core 30/Breast cancer module (EORTC QLQ-C30/BR23)
 - Godin Leisure-Time Exercise Questionnaire
 - Körperliche Aktivitäts-Skalen (Physical Activity Scales) developed by the Institute of Circulation Research and Sports Medicine at the Sport University Cologne, Germany
- The Kaplan-Meier method was used to calculate PFS estimates
- According to the methodologic features of an observational NIS, all statistical analyses were descriptive and the presented results should be interpreted as such
- Second interim analysis was planned to occur 12 months after inclusion of the 500th patient (data cut-off, 08 July 2014)
 - Data on baseline characteristics, PFS, tumor status, and safety from the first 500 documented patients were analyzed

Results

Study Status and Premature Discontinuation

Study Status
- 1,348 patients had entered study documentation through 19 August 2014
- This second interim analysis includes data for the first 500 patients (enrolled at 191 sites)
- The 500th patient had been included 12 months before the data cut-off on 8 July 2014

Premature Study Discontinuation
- At the time of data cut-off, 91 patients (18.2%) were still receiving treatment and 409 patients (81.8%) had documented discontinuation
- Reasons for study discontinuation were discontinuation of EVE+EXE therapy (67.0%), death (7.6%), consent withdrawal (3.4%), lost to follow-up (2.8%), and missing data (18.8% [including ongoing patients, 18.2%])
- Reasons for discontinuation of EVE+EXE are summarized in **Table 1**

Table 1. Reasons for Discontinuation of EVE+EXE

Discontinuation of Therapy With EVE+EXE[a]	Patients, n (%) (N = 500)
Progression	204 (40.8)
Adverse event	113 (22.6)
Poor compliance	4 (0.8)
Patient's wish	69 (13.8)
Missing (including ongoing patients)	110 (22.0)

[a]The data capture form asks for reason for discontinuation of documentation and reason for discontinuation of therapy as 2 separate questions. Therefore, various combinations of reasons are possible.

Baseline Patient and Disease Characteristics

- Median time from primary diagnosis was 7.2 years
 - Includes patients with first diagnosis in advanced setting
 - 20.8% of patients had metastases at primary diagnosis
- Median time from first diagnosis of recurrence/metastases was 2.7 years
- 97.6% of patients had metastatic disease at study entry
 - 53.7% of patients had visceral metastases at baseline
- 43.6% of patients had Eastern Cooperative Oncology Group (ECOG) performance status 0 at baseline
- 86.7% of patients received 10-mg EVE at the start of therapy
 - 13.3% received an EVE starting dose of 5 mg, which is half of the approved starting dose in this setting
- Additional baseline demographic and disease characteristics are reported in **Table 2**

Table 2. Baseline Patient and Disease Characteristics[a]

Patient Characteristics	Patients, n (%) (N = 500)
Age, median (range), years	66.0 (20.0, 93.0)
Weight, median, kg	70.0
Height, median, cm	165
Body mass index, median	25.9
ECOG Performance Status	**Patients, n (%) (n = 420)[a]**
0	183 (43.6)
1	194 (46.2)
2	39 (9.3)
3	3 (0.7)
4	1 (0.2)
Missing	80
Metastasis[b]	**Patients, n (%) (n = 495)[a]**
Visceral (lung, liver, CNS)	266 (53.7)
Visceral and bone	195 (39.4)
Visceral without bone	71 (14.3)
Only bone	130 (26.3)
Bone and other	250 (50.5)
Only other	110 (22.2)
Metastasis Location[c]	**Patients, n (%) (n = 495)[a]**
Lung	136 (27.5)
Bones	380 (76.8)
Liver	175 (35.4)
CNS	12 (2.4)
Skin	23 (4.6)
Lymph nodes	133 (26.9)
Other localization	76 (15.4)
No metastases	5 (1.0)
Hormone Receptor Status[d]	**Patients, n (%) (N=500)**
Estrogen receptor (ER)	
Positive	489 (98.6)
Negative	7 (1.4)
Unknown	0
Missing	4
Progesterone receptor (PgR)	
Positive	383 (77.2)
Negative	107 (21.6)
Unknown	6 (1.2)
Missing	4
Combination of ER and PgR	
ER$^+$/PgR$^+$	376 (75.8)
ER$^+$/PgR$^-$	107 (21.6)
ER$^-$/PgR$^+$	7 (1.4)
> 1 unknown value	6 (1.2)
Missing	4
Ki-67 Index[d]	**Patients, n (%) (N=500)**
< 10%	31 (7.8)
10%-14%	40 (10.1)
15%-20%	47 (11.8)
> 20%	71 (17.8)
Unknown	209 (52.5)
Missing	102

[a] Note that percentages are based on number of patients with available data and numbers of patients are reported in the column headings.
[b] Percentages add to >100% because the categories listed are not mutually exclusive.
[c] Percentages add to >100% because the same patient could have >1 metastasis location.
[d] Hormone receptor status and Ki-67 index are calculated based on the number of patients with available data.
Abbreviations: CNS, central nervous system; ECOG, Eastern Cooperative Oncology Group.

EVE+EXE Treatment Characteristics

- More than half of patients received EVE+EXE as first-line or second-line treatment for advanced disease (**Figure 2**)
 - Most patients (445, 89.0%) switched to EVE+EXE because of disease progression on a previous therapy
- Among patients receiving EVE+EXE in the advanced setting
 - First- or second-line therapy: letrozole and anastrozole were the most common last prior antineoplastic therapy
 - Third- or later-line therapy: fulvestrant or chemotherapy were increasingly documented as last prior therapy
 - 93 patients (18.6%) had received EXE as prior antineoplastic therapy (6% as last antineoplastic therapy)

Figure 2. Distribution of line of therapy for EVE+EXE (N = 500).

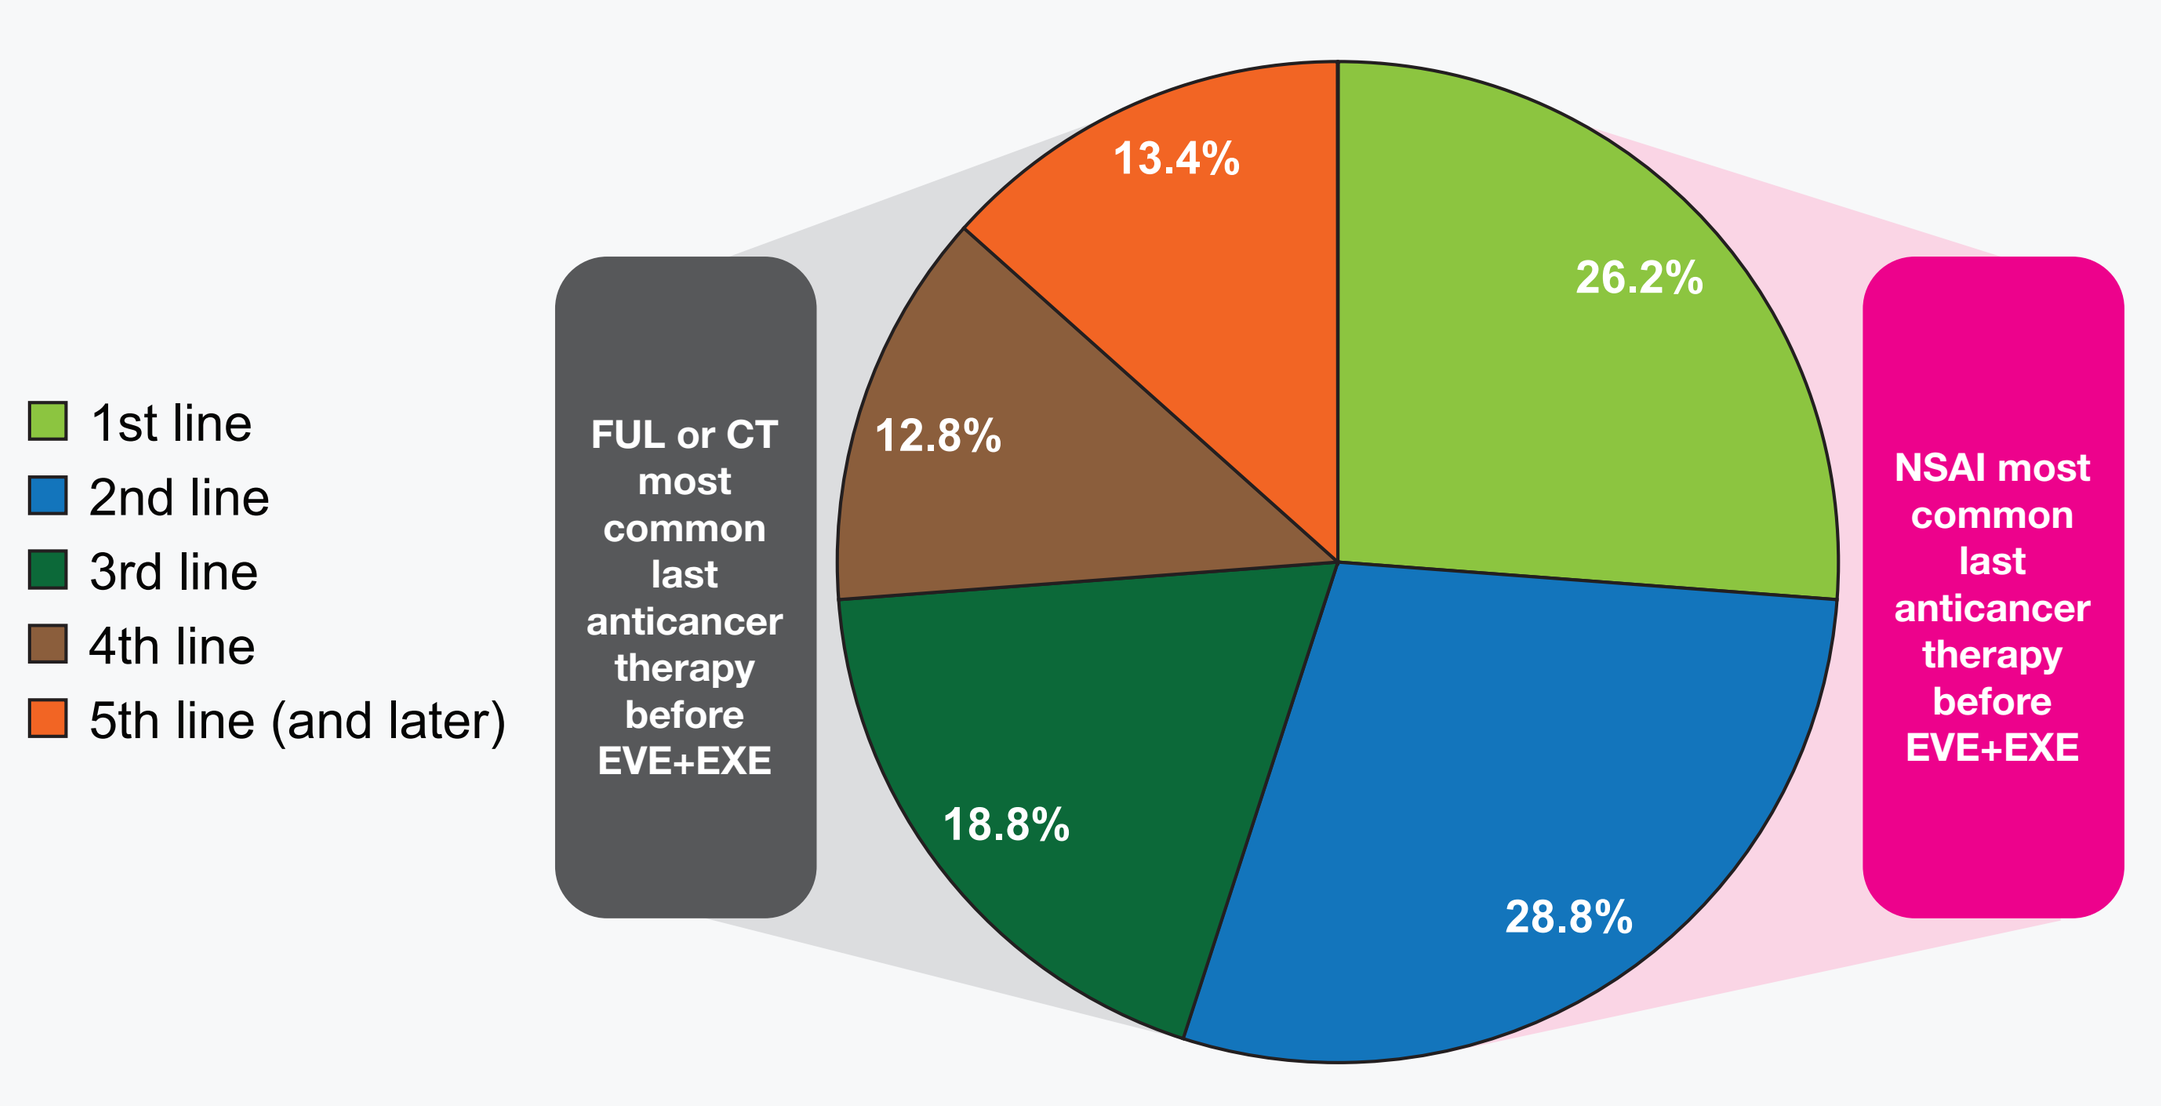
1st line
2nd line
3rd line
4th line
5th line (and later)
FUL or CT most common last anticancer therapy before EVE+EXE
13.4%
26.2%
12.8%
18.8%
28.8%
NSAI most common last anticancer therapy before EVE+EXE

Abbreviations: CT, chemotherapy; EVE, everolimus; EXE, exemestane; FUL, fulvestrant; NSAI, nonsteroidal aromatase inhibitor.

Progression-Free Survival

- Median PFS for the first 500 patients was 8.0 months (95% confidence interval [CI], 6.7-9.1; **Figure 3**)

Figure 3. Progression-free survival (first 500 patients).

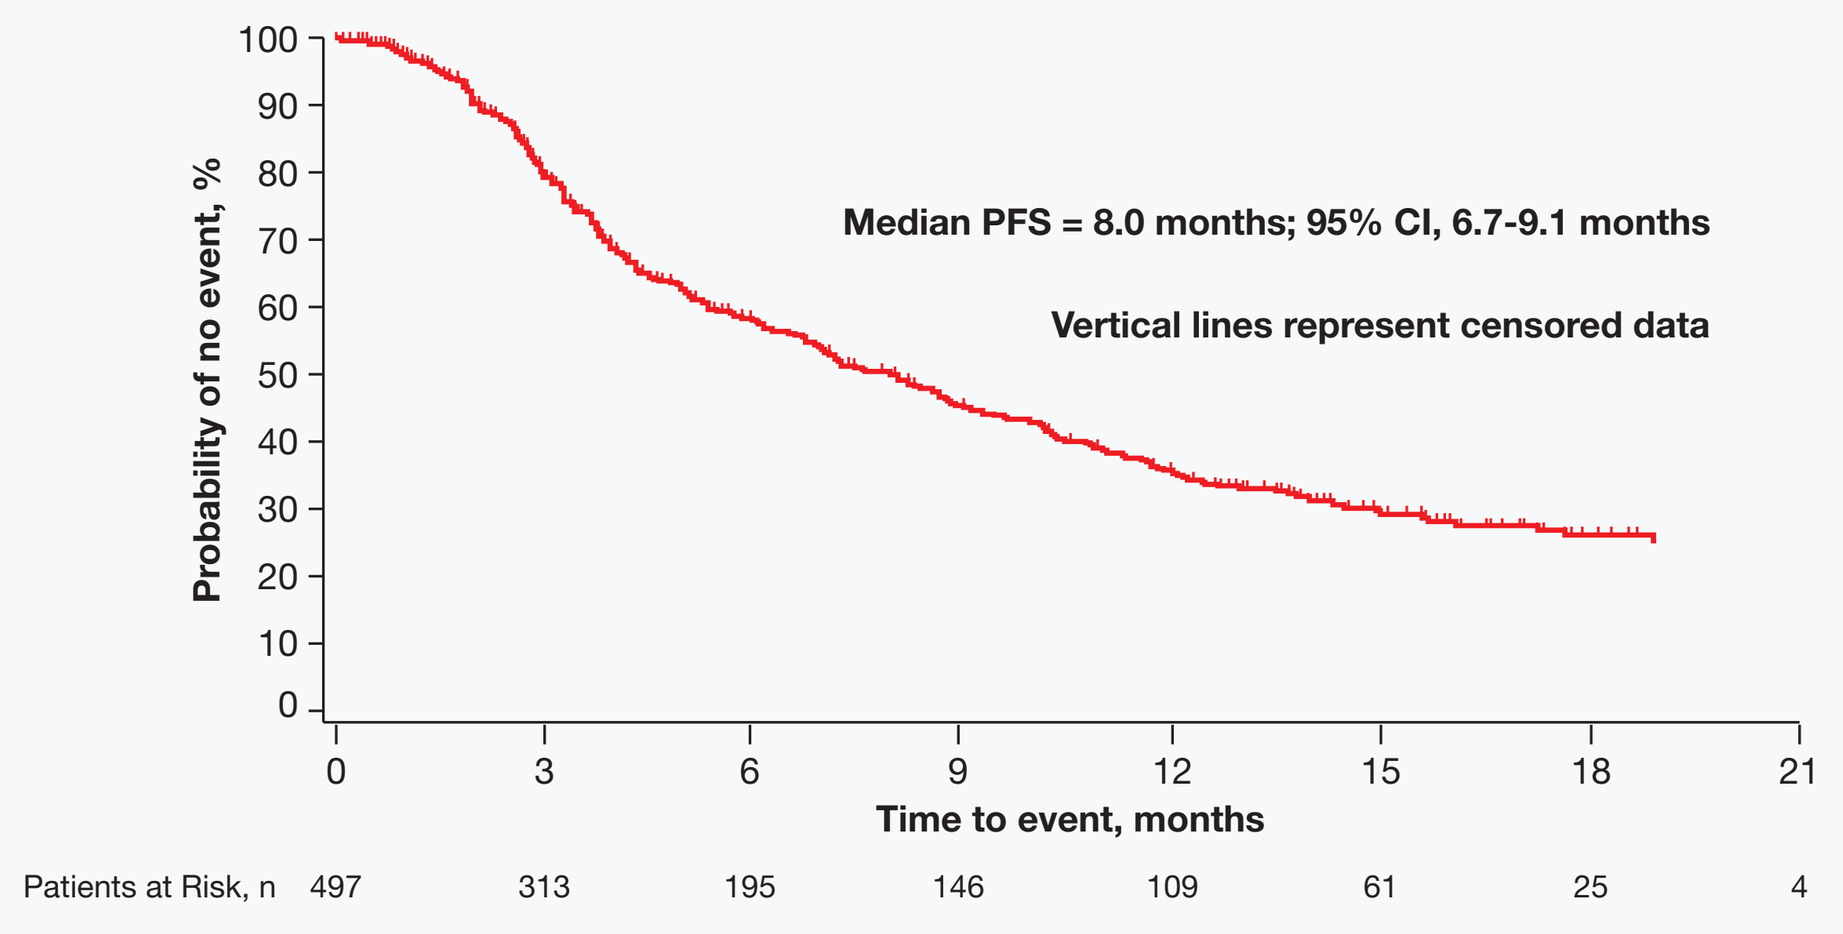
Probability of no event, %
100
90
80
70
60
50
40
30
20
10
0
Median PFS = 8.0 months; 95% CI, 6.7-9.1 months
Vertical lines represent censored data
0 3 6 9 12 15 18 21
Time to event, months
Patients at Risk, n 497 313 195 146 109 61 25 4

Abbreviations: CI, confidence interval; PFS, progression-free survival.

Overall Response Rate

- Patients treated with EVE+EXE had an overall response rate (ORR) of 19.4% (complete response, 0.7%; partial response, 18.7%)

Progression-Free Survival—Subgroup Analyses

- Median PFS was longer when EVE+EXE was administered as first- or second-line therapy compared with later-line therapy (**Figure 4**)
- Median PFS was comparable for patients with or without prior exemestane treatment (**Figure 5**)
- Median PFS in additional patient subgroups of interest is presented in **Table 3**
 - Median PFS was longer in the subgroup of patients with bone-only metastases than in the subgroup with visceral metastases, which is consistent with previous observations from BOLERO-2[2]

Figure 4. Progression-free survival by line of therapy (n = 497).

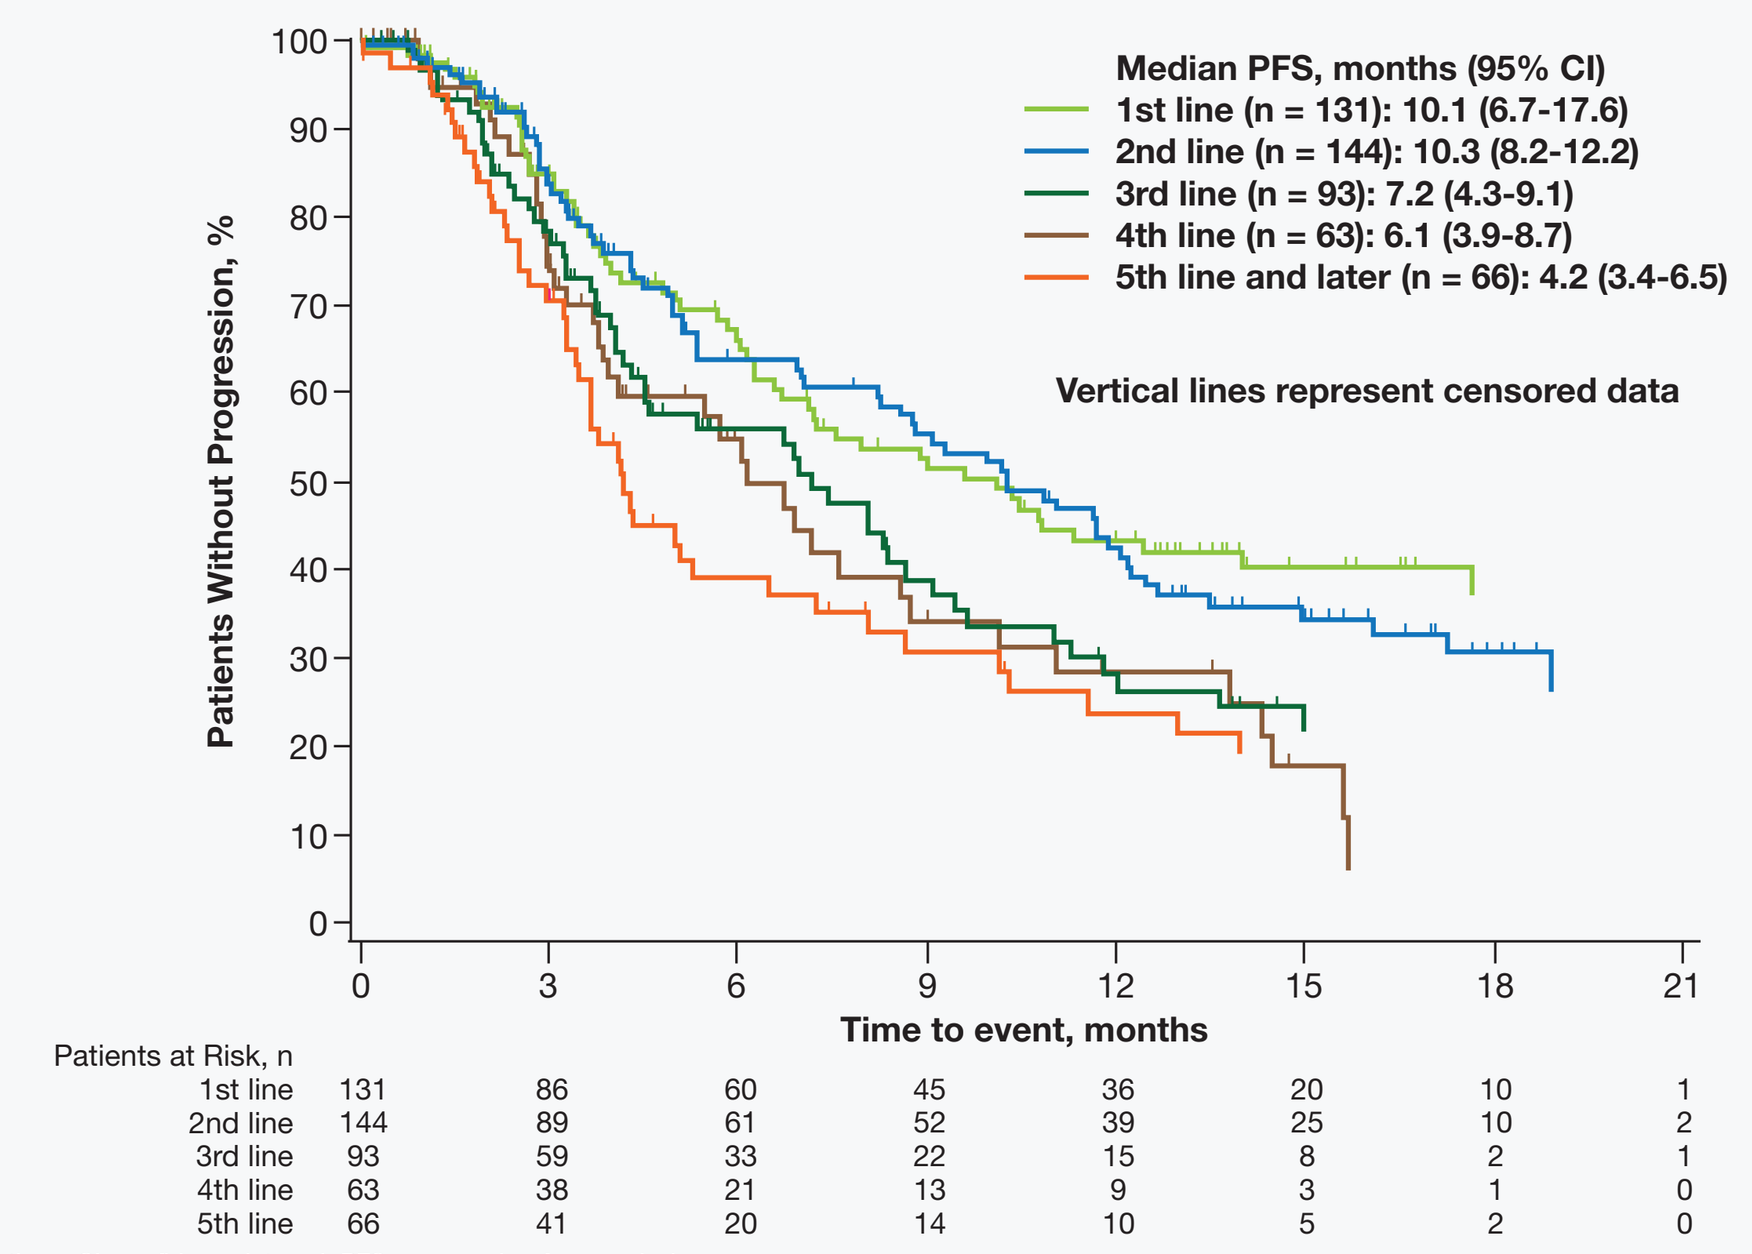
Patients Without Progression, %
100
90
80
70
60
50
40
30
20
10
0
Median PFS, months (95% CI)
1st line (n = 131): 10.1 (6.7-17.6)
2nd line (n = 144): 10.3 (8.2-12.2)
3rd line (n = 93): 7.2 (4.3-9.1)
4th line (n = 63): 6.1 (3.9-8.7)
5th line and later (n = 66): 4.2 (3.4-6.5)
Vertical lines represent censored data
0 3 6 9 12 15 18 21
Time to event, months
Patients at Risk, n
1st line 131 86 60 45 36 20 10 1
2nd line 144 89 61 52 39 25 10 2
3rd line 93 59 33 22 15 8 2 1
4th line 63 38 21 13 9 3 1 0
5th line 66 41 20 14 10 5 2 0

Abbreviations: CI, confidence interval; PFS, progression-free survival.

Figure 5. Progression-free survival by prior exemestane (n = 497).

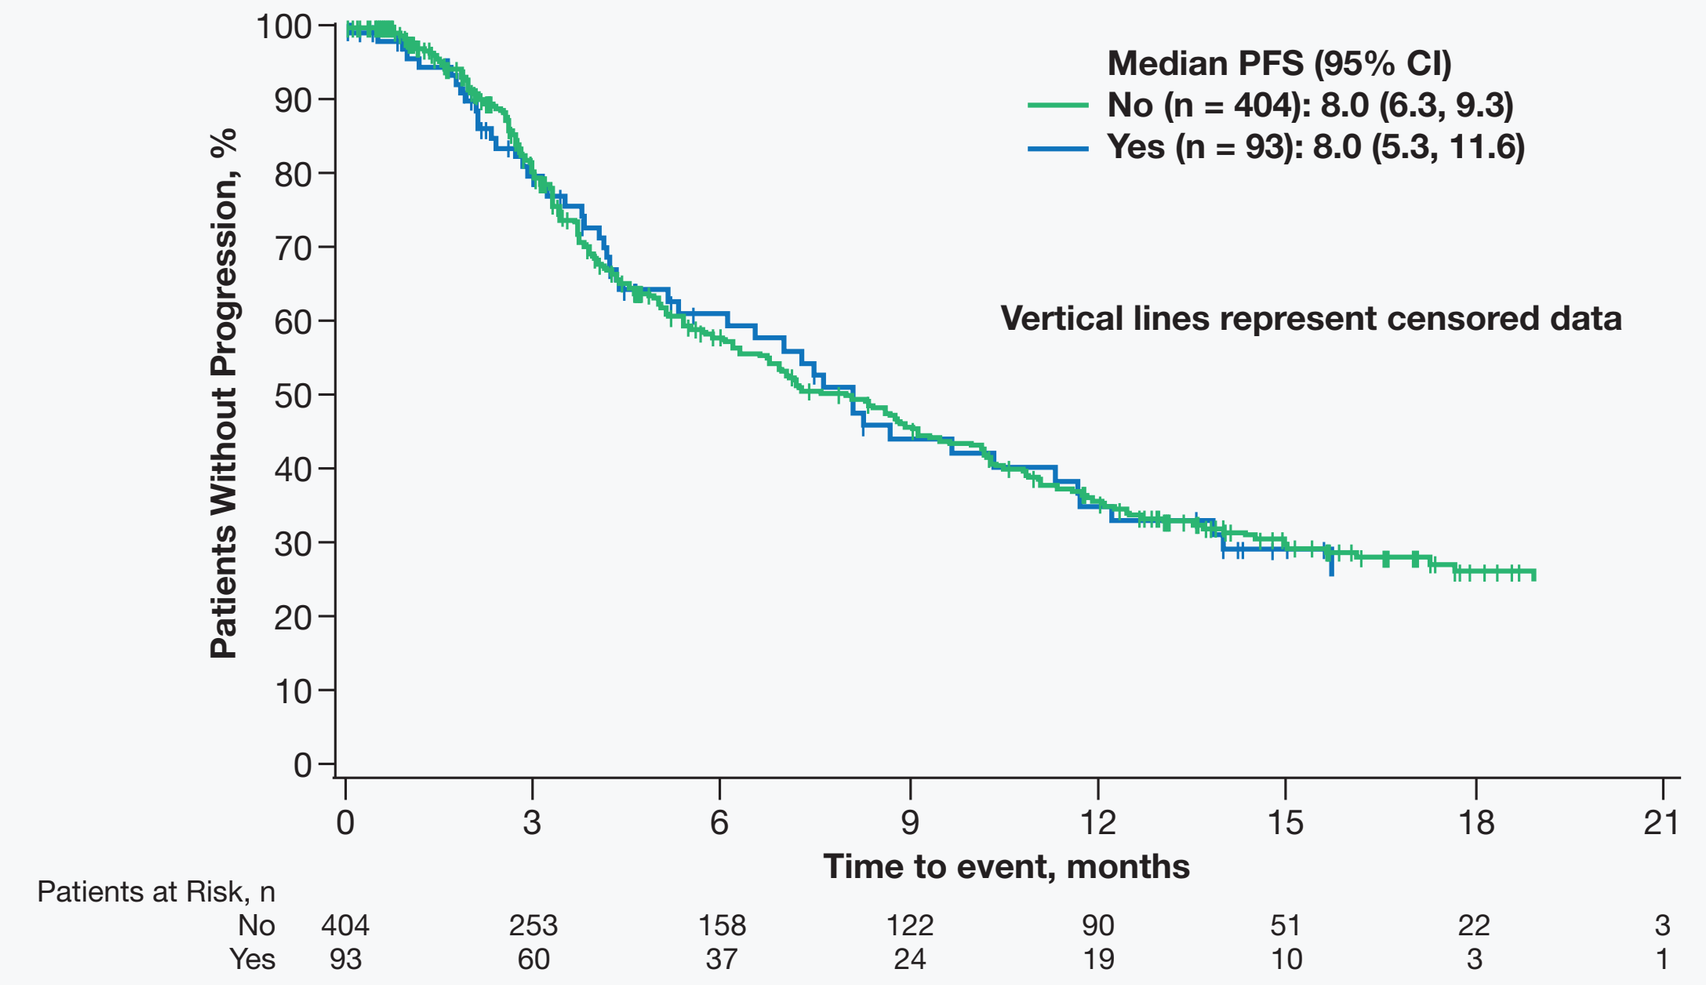
Patients Without Progression, %
100
90
80
70
60
50
40
30
20
10
0
Median PFS (95% CI)
No (n = 404): 8.0 (6.3, 9.3)
Yes (n = 93): 8.0 (5.3, 11.6)
Vertical lines represent censored data
0 3 6 9 12 15 18 21
Time to event, months
Patients at Risk, n
No 404 253 158 122 90 51 22 3
Yes 93 60 37 24 19 10 3 1

Abbreviations: CI, confidence interval; PFS, progression-free survival.

Table 3. Progression-Free Survival—Additional Subgroup Analyses

	Patients, n	Median, months, (95% CI)
Prior chemotherapy in the advanced setting[a]		
No	333	8.8 (7.1, 10.8)
Yes	164	6.1 (4.3, 8.3)
Sensitivity to prior hormonal therapy[b]		
Yes	333	7.2 (5.5, 9.0)
No	150	8.0 (6.2, 10.3)
Ki-67 expression (assessed at baseline)		
≤ 20%	118	8.7 (6.7, 11.3)
> 20%	70	4.3 (3.3, 6.9)
Bone and visceral metastases[c]		
Bone only	130	10.9 (8.6, NR)
Single visceral metastasis ± bone	209	6.3 (4.5, 7.6)
Multiple visceral metastases ± bone	54	4.1 (3.3, 6.1)
Disease progression[d]		
1st-line with rapid progression[e]	54	10.4 (6.0, NR)
1st-line with longer recurrence-free interval[f]	29	14.0 (4.0, NR)
Stomatitis (Within 8 weeks of start of EVE)		
Yes	219	8.3 (6.6, 10.2)
No	278	7.2 (6.0, 9.4)
Physical activity[g]		
Active patients	63	10.2 (7.6, 13.8)
Moderately active patients	53	5.7 (4.0, 12.6)
Insufficiently active patients	222	7.4 (6.0, 9.0)

[a]Analysis only includes chemotherapy for advanced disease.
[b]Patients who received > 24 months of adjuvant hormonal therapy prior to recurrence. Patients with best response defined as stable disease for > 168 days in the advanced setting. This definition is similar to the BOLERO-2 definition of endocrine sensitivity[5]
[c]94 patients had skin, lymph node, or other metastases without visceral involvement; for 5 patients metastatic disease had not been documented; data for 5 patients are missing.
[d]Data regarding recurrence-free interval were not interpretable for 48 patients
[e]Recurrence ≤ 12 months after adjuvant therapy.
[f]Recurrence > 12 months after adjuvant therapy.
[g]Based on Godin Leisure-Time Exercise Questionnaire (assessed at baseline).
Abbreviation: CI, confidence interval; NR, not reached.

Safety

- Adverse events (AEs; **Table 4**) were consistent with those previously reported with EVE+EXE in patients with advanced breast cancer[1]
- Most commonly reported AEs of any grade in ≥ 10% of patients
 - Stomatitis (39.8%)
 - Fatigue (15.6%)
 - Diarrhea (13.2%)
 - Dyspnea (13.0%)
 - Nausea (12.0%)
 - Decreased appetite (10.4%)

Table 4. Adverse Events

Adverse Event (Preferred Term)	Patients With Adverse Event by Grade, n (%)[a,b] (N = 500)				
	Any grade	Grade 1	Grade 2	Grade 3	Grade 4
Stomatitis	199 (39.8)	116 (23.2)	85 (17.0)	17 (3.4)	1 (0.2)
Fatigue	78 (15.6)	43 (8.6)	28 (5.6)	11 (2.2)	0
Diarrhea	66 (13.2)	29 (5.8)	40 (8.0)	4 (0.8)	0
Dyspnea	65 (13.0)	24 (4.8)	20 (4.0)	14 (2.8)	3 (0.6)
Nausea	60 (12.0)	26 (5.2)	26 (5.2)	9 (1.8)	1 (0.2)
Decreased appetite	52 (10.4)	22 (4.4)	25 (5.0)	4 (0.8)	0
Peripheral edema	49 (9.8)	28 (5.6)	16 (3.2)	4 (0.8)	0
Cough	44 (8.8)	22 (4.4)	18 (3.6)	2 (0.4)	0
General physical health deterioration	44 (8.8)	6 (1.2)	17 (3.4)	16 (3.2)	4 (0.8)
Malignant neoplasm progression	43 (8.6)	0	5 (1.0)	9 (1.8)	18 (3.6)
Dysgeusia	38 (7.6)	22 (4.4)	14 (2.8)	2 (0.4)	0
Rash	38 (7.6)	20 (4.0)	20 (4.0)	0	0
Epistaxis	35 (7.0)	27 (5.4)	8 (1.6)	1 (0.2)	0
Weight decreased	34 (6.8)	14 (2.8)	18 (3.6)	2 (0.4)	0
Vomiting	33 (6.6)	13 (2.6)	12 (2.4)	7 (1.4)	0
Arthralgia	31 (6.2)	21 (4.2)	9 (1.8)	2 (0.4)	0
Headache	30 (6.0)	20 (4.0)	10 (2.0)	1 (0.2)	0
Anemia	30 (6.0)	10 (2.0)	18 (3.6)	4 (0.8)	0
Pruritus	28 (5.6)	17 (3.4)	9 (1.8)	1 (0.2)	0
Pain in extremity	26 (5.2)	14 (2.8)	10 (2.0)	3 (0.6)	0
Nasopharyngitis	25 (5.0)	15 (3.0)	11 (2.2)	1 (0.2)	0
Pleural effusion	25 (5.0)	1 (0.2)	12 (2.4)	6 (1.2)	3 (0.6)
Pyrexia	25 (5.0)	10 (2.0)	9 (1.8)	4 (0.8)	0

[a]Adverse events in ≥5% of patients.
[b]Grade 1 = mild; grade 2 = moderate; grade 3 = severe; grade 4 = life-threatening.

- 174 patients discontinued treatment because of an AE
 - Stomatitis (12.6%), general physical health deterioration (9.8%), dyspnea (6.9%), pleural effusion (6.3%), fatigue (5.7%), and nausea (5.2%) were the most common AEs resulting in discontinuation

Everolimus Dose Intensity and Treatment Compliance

- EVE was administered at a median relative dose intensity of 88.1% (n = 497)
 - Patients starting at 10.0 mg (n = 431) received a median dose intensity of 93.5%, whereas patients starting at 5.0 mg (n = 66) received a considerably lower median dose intensity of 50.0%
- Based on physician's assessment of patient diaries (recommended but not mandatory), most patients were compliant with EVE therapy
 - During the first 12 months of therapy, estimated intake of EVE was between 80% and 100% of the intended dose in > 90% of patients

Follow-up Therapy

- Among 410 patients who completed EVE+EXE therapy, follow-up was planned in 270 (67.8%) patients (no follow-up planned in 119 [29%] patients)
 - 178 (43.4%) patients received chemotherapy as follow-up treatment, and later-line patients were more likely to receive chemotherapy than earlier-line patients
 - 92 patients (22.4%) received endocrine therapy and 7 patients (1.7%) received experimental therapy

Discussion

- Median PFS in BRAWO was consistent with the overall population in BOLERO-2 (8.0 months vs 7.8 months, respectively)[1] and in patients receiving EVE+EXE as first-line therapy in the advanced setting (10.1 months vs 11.5 months, respectively)[3]
- Patients receiving EVE+EXE as 1st- or 2nd-line treatment had a longer PFS duration than those receiving later lines, regardless of EXE pretreatment or sensitivity to prior hormonal therapy
- Longer median PFS durations were also seen in the following patient subgroups:
 - Ki-67 expression ≤ 20%
 - Bone-only metastases
 - EVE+EXE as first-line therapy with longer recurrence-free interval
 - No prior chemotherapy
- For the first time, data are available demonstrating that PFS for patients with EXE pretreatment is comparable to PFS for patients without EXE pretreatment
 - Patients with EXE pretreatment were excluded from BOLERO-2
- The overall safety profile in the BRAWO study is comparable to that observed in BOLERO-2[1] and reported for EVE in other cancer indications[4]
 - The percentage of patients with any grade stomatitis was lower in BRAWO (39.8%) than in BOLERO-2 (59%)
- Estimated intake of EVE was between 80% and 100% of the intended dose in > 90% of patients according to patient diaries

Acknowledgments

The results are presented on behalf of all sites participating in the BRAWO study.
We thank the patients for their participation in this non-interventional study, as well as the staff at each documenting site and Winicker Norimed GmbH (Nuremberg, Germany), all of whom significantly contributed to the success of the study. Michael Albrecht, Novartis Pharma GmbH Nuremberg, provided a significant contribution to all questions regarding pharmacovigilance. Medical editorial and graphics assistance for this poster presentation was supported by Novartis Pharmaceuticals Corporation.

References

1. Yardley DA, et al. *Adv Ther.* 2013;30:870-884.
2. Campone M, et al. *Eur J Cancer.* 2013;49:2621-2632.
3. Beck T, et al. *Breast Cancer Res Treat.* 2014;143:459-467.
4. Afinitor 2014 Summary of Product Characteristics.
5. Baselga J, et al. *N Engl J Med.* 2012;366:520-529.

Presented at SABCS 2014 Congress; 9-13 December 2014; San Antonio, Texas, USA. Abstract P5-19-12.

Supported by

NOVARTIS ONCOLOGY

Arbeitskreis Klinische Studien in onkologischen und hämatologischen Praxen e.V.

Deutsche Gesellschaft für Senologie

Deutsche Sporthochschule Köln
German Sport University Cologne
Institut für Kreislaufforschung und Sportmedizin